The epicutaneous immunotherapy company

No Impact of Filaggrin Deficiency on EPIT® in a Murine Model

Wavrin S. [1], Mondoulet L. [1], Dioszeghy V. [1], Plaquet C. [1], Puteaux E. [1], Ligouis M.[1], Dhelft V. [1], Prof. Dupont C. [1], Benhamou P.-H [1]

1. DBV Technologies, Montrouge, France

Rationale: Epicutaneous immunotherapy (EPIT), currently investigated in the treatment of food allergy, needs the integrity of the skin to warrant safety and efficacy. Mutations in the gene encoding the key epidermal protein filaggrin (FLG) are risk factors for peanut allergy and disrupt the skin integrity. We investigated the association between FLG deficiency and peanut EPIT efficacy in a murine model.

Method: FLG mutant mice deficient in filaggrin (FLG-/-) or wild-type (WT) mice were sensitized with peanut protein extract (peanut protein) and cholera toxin. Sensitized mice received one patch per week for 8 weeks of EPIT, using Viaskin, and were then subjected to sustained oral peanut exposure. We assessed blood humoral and cellular responses, and evaluated eosinophil infiltration in the esophageal mucosa after peanut sustained regimen. The different steps of allergen capture and transportation following deposition on the skin were also analyzed in sensitized mice.

Results: Sensitization of mice was confirmed by a significant increase of specific Th2 biased immunological responses. In sensitized mice, EPIT significantly reduced IgE levels, splenocytes secretion of Th2 cytokines and recruitment of eosinophils in the esophagus compared to WT mice. The allergen applied onto the skin of FLG-/- mice did not undergo passive skin passage or systemic delivery. Instead, the allergen was captured by skin CD205high DCs, which migrated to afferent lymph nodes, as already described in WT mice.

Conclusion: EPIT was efficient and safe in FLG-/- mice, suggesting that in humans, EPIT will be efficacious and safe in the presence of FLG polymorphism or mutations.